Current consumption behavior, burden and need of support among Austrian MSM practicing chemsex

Birgit Leichsenring[1], Thomas Baumgartner[2], Matthias Egger[3]

[1]medinfo.at, Vienna, Austria, [2]Aids Hilfe Wien, Vienna, Austria, [3]Gilead Sciences GesmbH, Vienna, Austria

Poster 36

DÖAK 2025

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Background

The intentional use of illicit substances (chems) among men who have sex with men (MSM) in a sexual context (chemsex), has been reported a substantial public health issue.[1-3] To pool expertise and offer professional support, a nation-wide network of experts from different professions related to all aspects of chemsex has been founded.[4]

This study aimed to assess consumption behavior, possible changes over time, burden, and need of support among Austrian MSM taking chems.

Methods

Austrian MSM were asked to participate in an online survey with closed-ended questions from May to October 2024. Four major drugs associated with chemsex, i.e. crystal methamphetamine, GHB/GBL, ketamine, and mephedrone, were included in the survey.[1-3] Volunteers were randomly recruited and excluded if chemsex experience was denied.

Results

A total of 102 users participated in the survey (**Figure 1**). As shown in **Figure 2**, the most frequently used substances are mephedrone and GHB/GBL followed by ketamine and crystal methamphetamine (92%, 86%, 61% and 47% of users). 25% consume at least weekly (**Figure 3**), 25% report experience with intravenous application (**Figure 4**), and 27% use chems also alone without sex (**Figure 5**).

54% started consumption within the last 5 years and increased use over time was reported for all 4 substances and greatest among mephedrone users (48%) (**Figure 6**).

62% estimate to have their use under control, 12% take or have already taken professional help (**Figure 7**). 61% know the website www.chemsex.at listing professional support services offered by the Austrian Chemsex Network.

Interpretation

Chemsex risks seem to be highly underestimated among Austrian users. The COVID-19 pandemic might have aggravated the problem over the past years. These findings strongly suggest an increasing need for education- and prevention measures as well as support services for users.

Figure 1. Age distribution and residence of participants

The majority of the 102 participants were in their thirties or forties (73%) and lived in the Vienna metropolitan region (75%). Previous surveys from the Austrian Chemsex Network with MSM practicing chemsex showed a similar distribution of age and residence.[5-6]

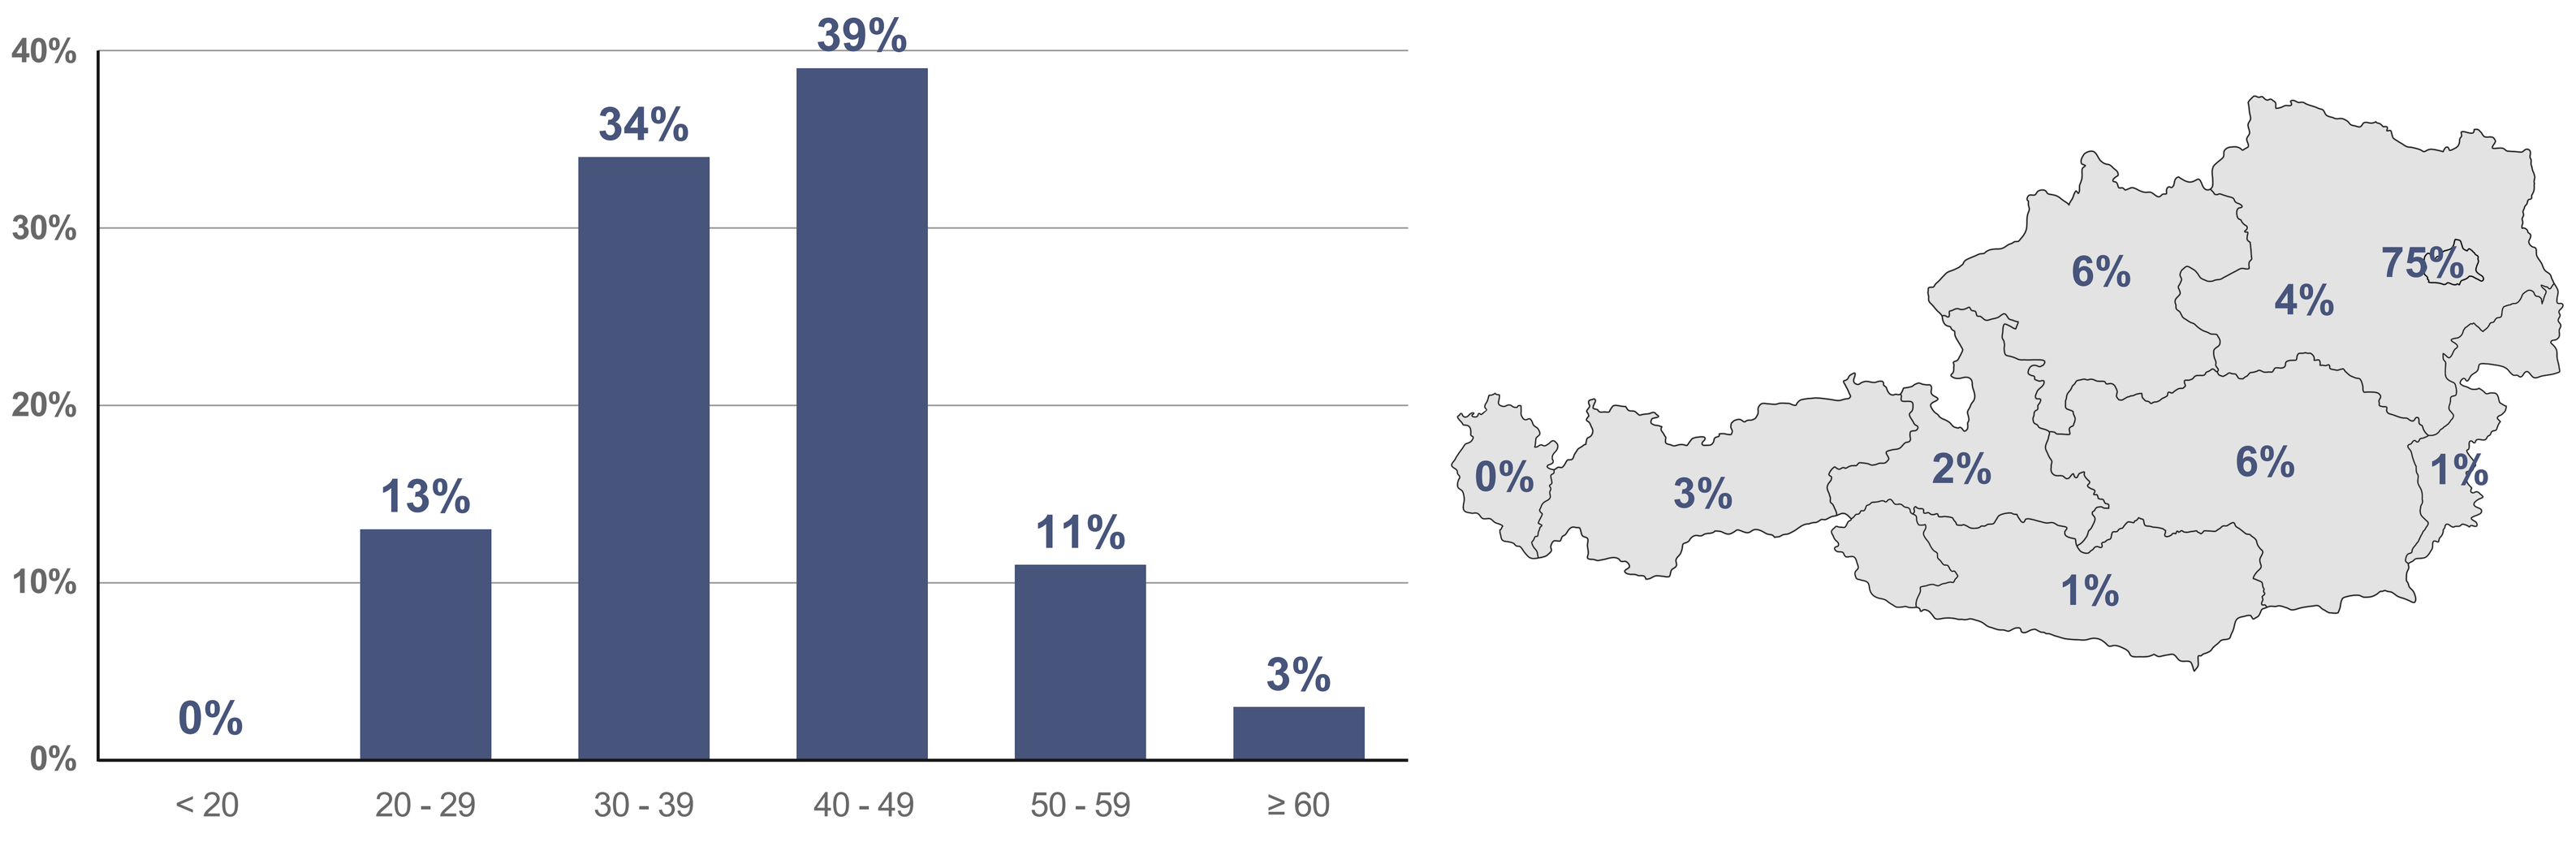
40%
30%
20%
10%
0%
0%
13%
34%
39%
11%
3%
< 20
20 - 29
30 - 39
40 - 49
50 - 59
≥ 60
0%
3%
2%
6%
4%
75%
6%
1%
1%

Figure 2. Substances used

Reporting all out of the 4 chemsex-associated substances studied, that the participants consume or have consumed at least once, showed the lowest use of crystal methamphetamine. Nevertheless, almost every second participant stated to have experience with this drug.

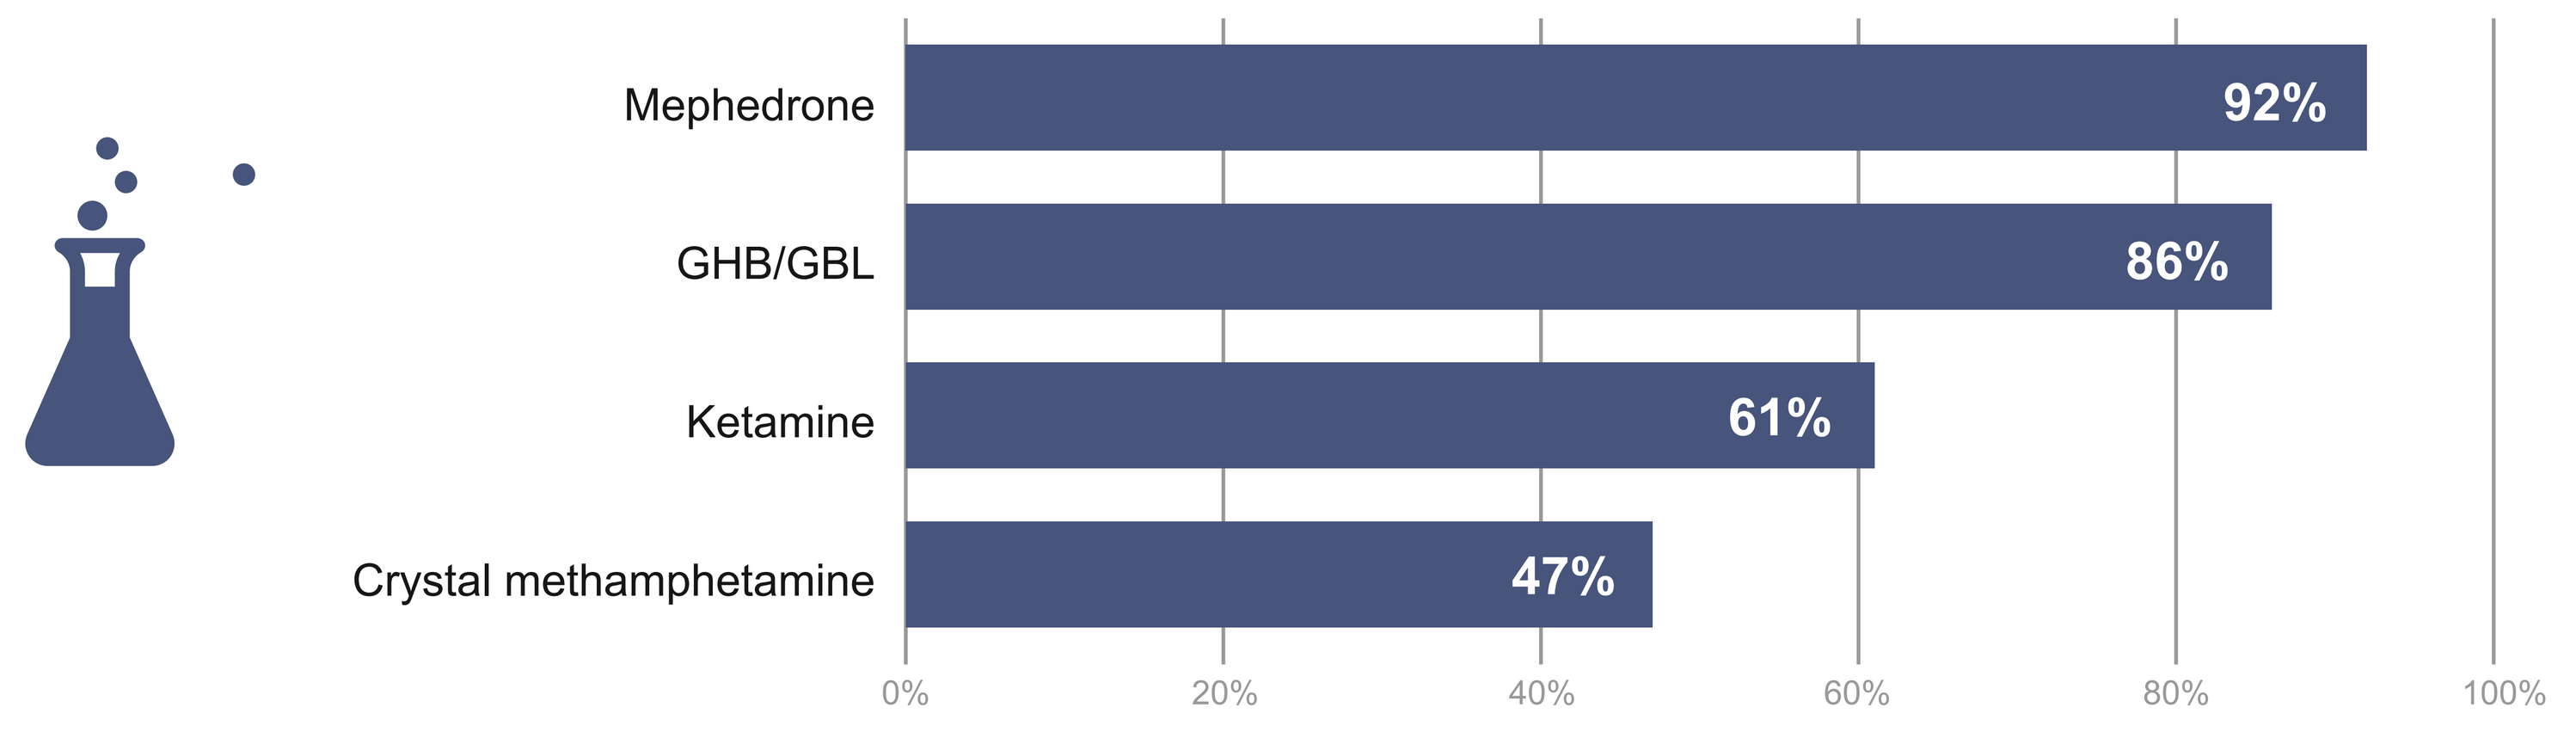
Mephedrone 92%
GHB/GBL 86%
Ketamine 61%
Crystal methamphetamine 47%
0% 20% 40% 60% 80% 100%

Figure 3. Consumption frequency

Distribution among consumption intervals was rather even. Every 4th participant reported weekly consumption: 16% once and 9% several times per week.

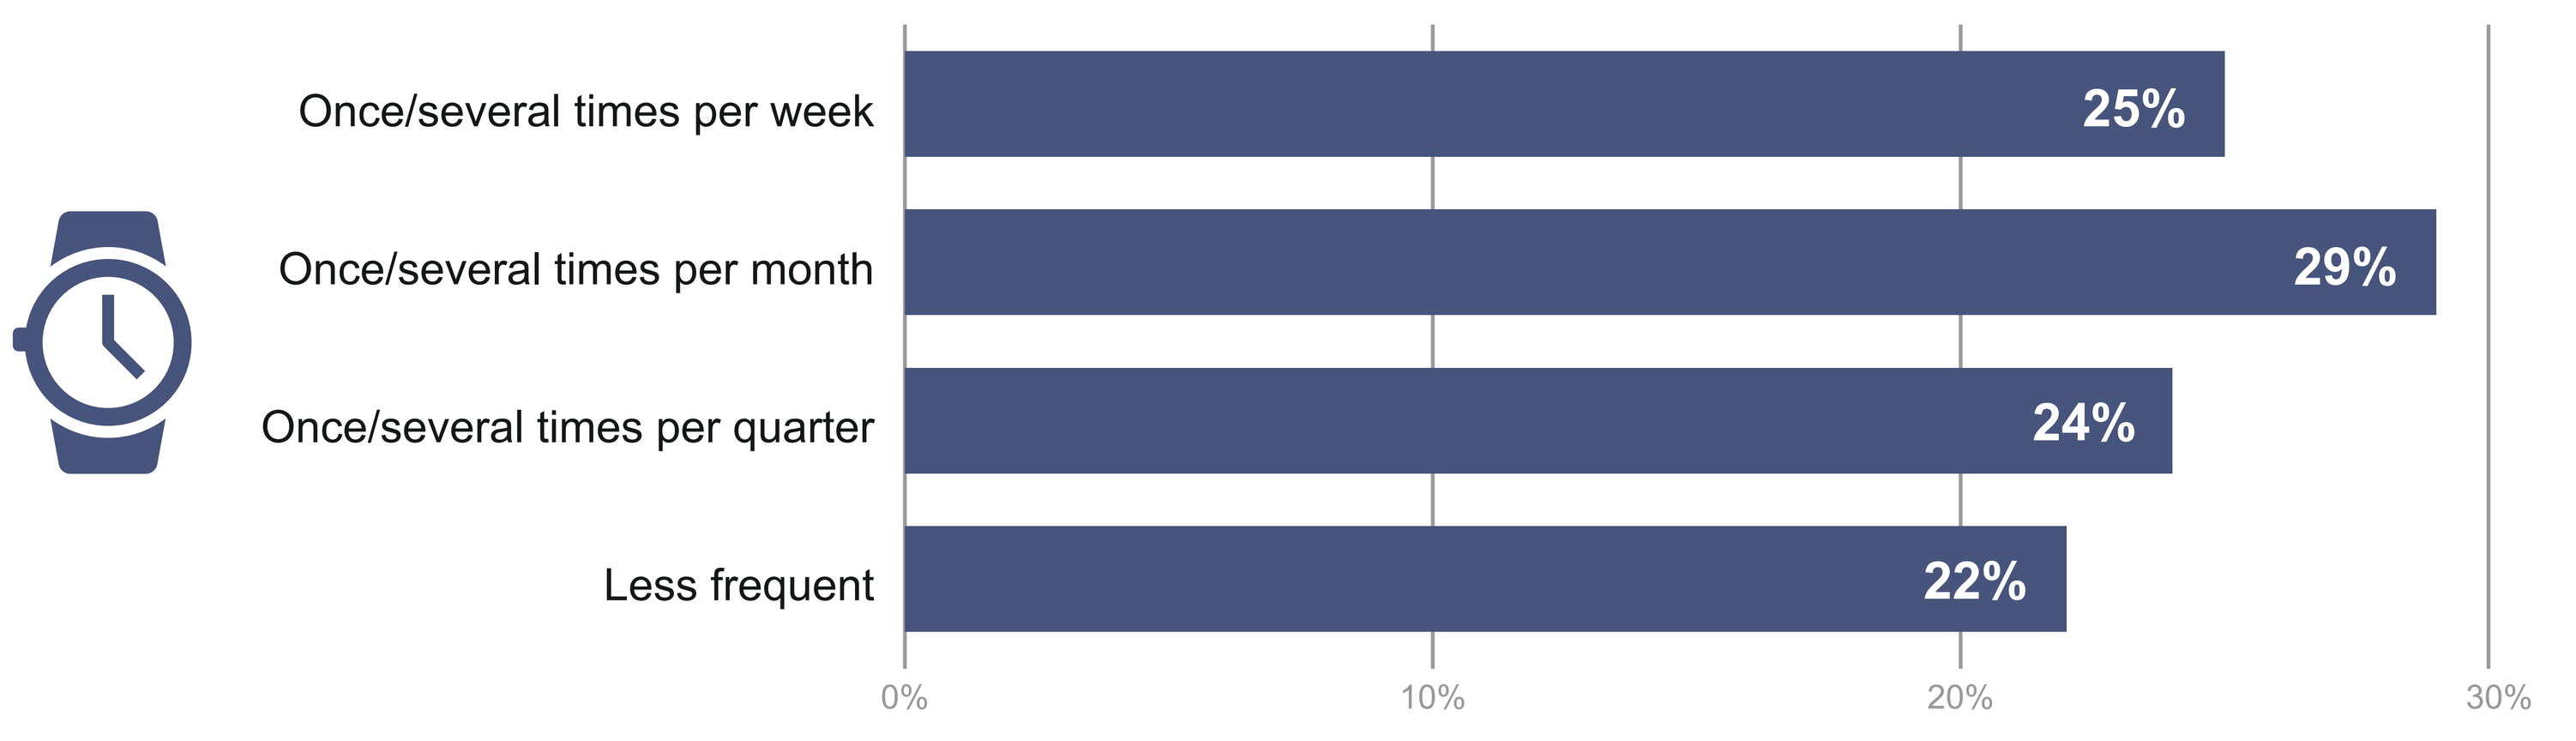
Once/several times per week 25%
Once/several times per month 29%
Once/several times per quarter 24%
Less frequent 22%
0% 10% 20% 30%

Figure 4. Routes of application

Sniffing (96% of participants) and drinking (86% of participangs) were the most frequent modes of application. However, one out of 4 participants also reported experience with intranvenous application („slamming“), 4% even on a regular base.

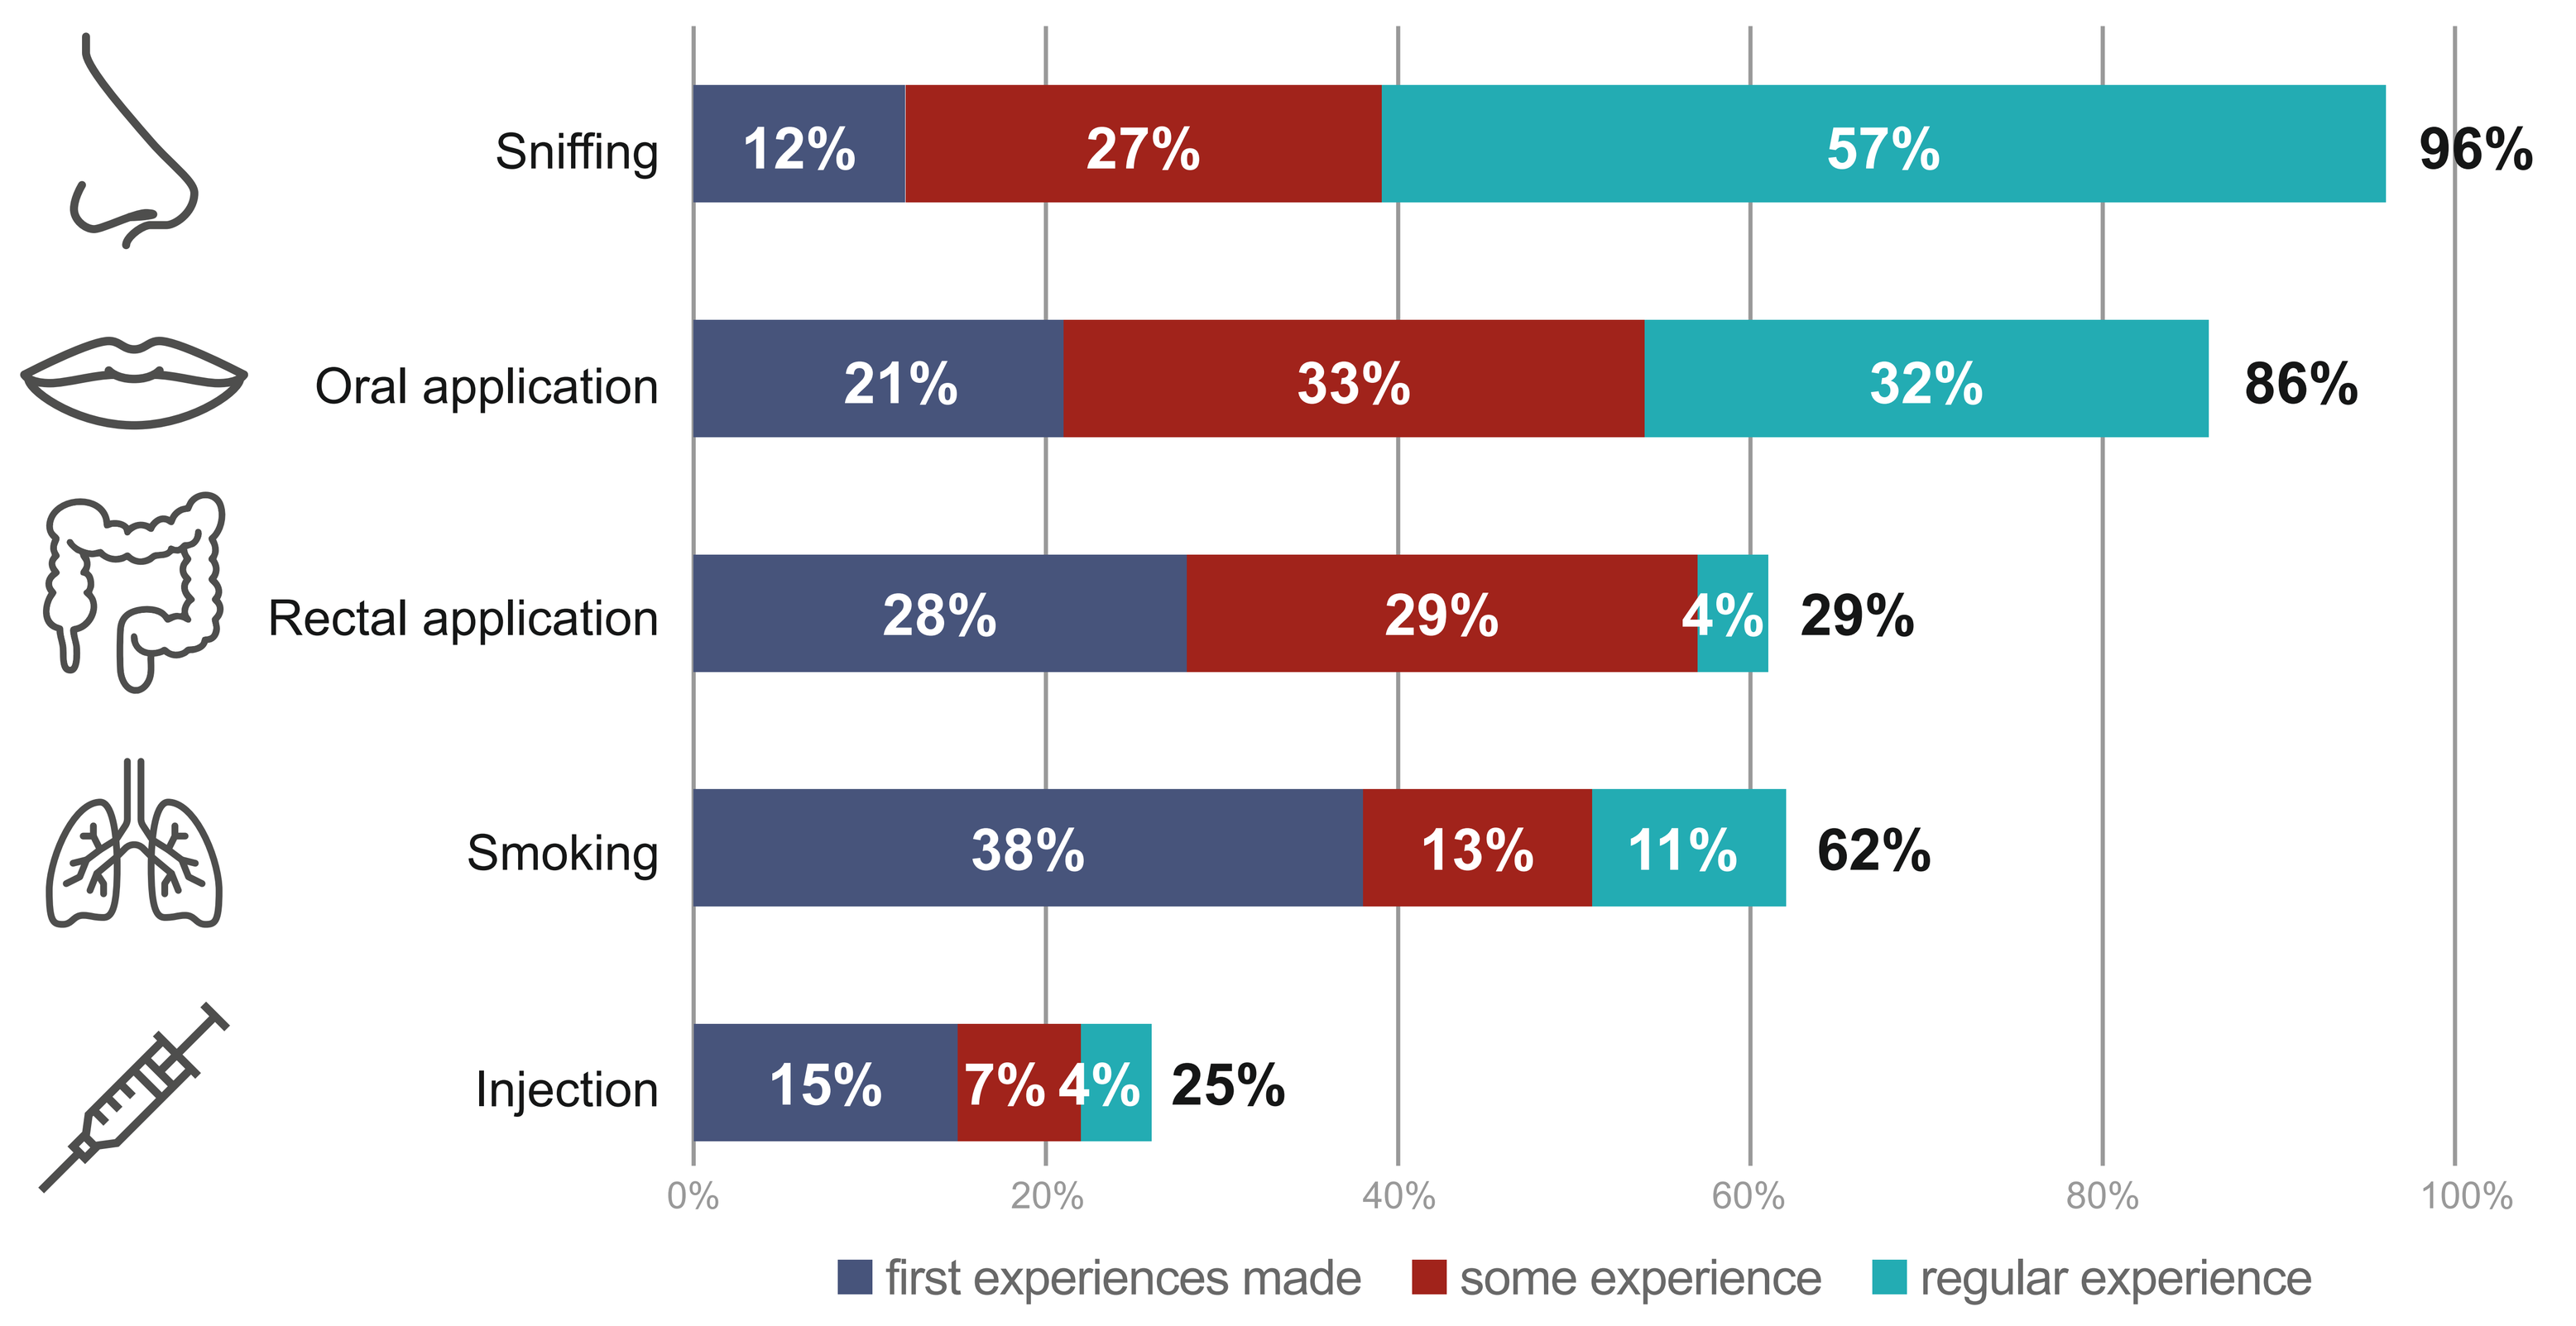
Sniffing 12% 27% 57% 96%
Oral application 21% 33% 32% 86%
Rectal application 28% 29% 4% 29%
Smoking 38% 13% 11% 62%
Injection 15% 7% 4% 25%
0% 20% 40% 60% 80% 100%
first experiences made
some experience
regular experience

Figure 5. Setting and context of consumption

The majority of participants reported consumption in a sexual context with other persons (97%) but a notable proportion also stated to consume in a solitary setting, either in a sexual context (51%) or without sex (27%).

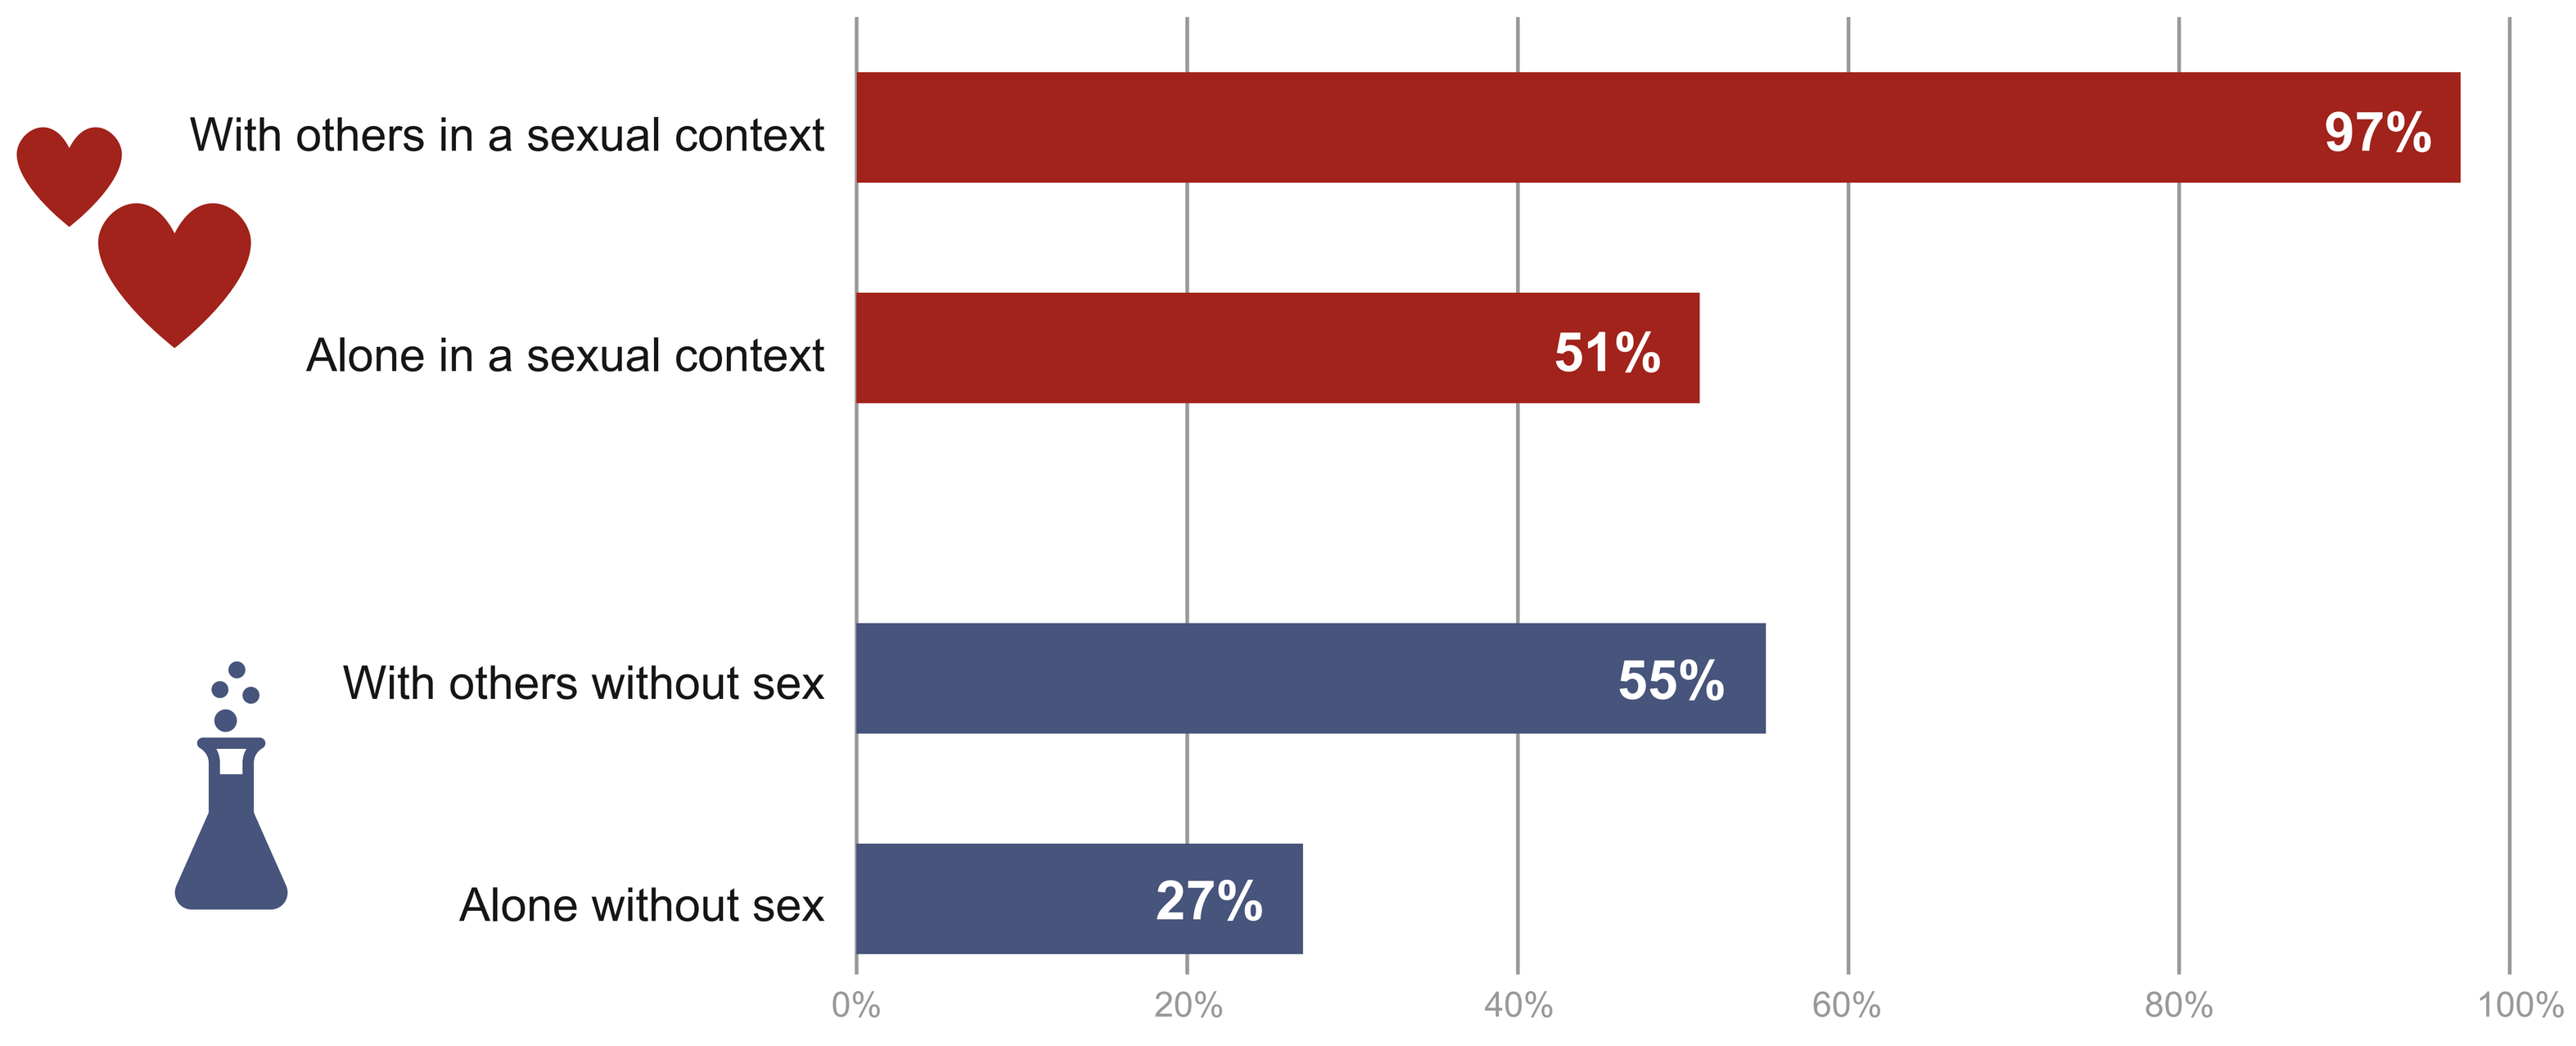
With others in a sexual context 97%
Alone in a sexual context 51%
With others without sex 55%
Alone without sex 27%
0% 20% 40% 60% 80% 100%

Figure 6. Time of first chems-experience and change in consumer behavior over time

More than half of the participants (54%) started consumption within the past 5 years. From those starting within the last year, 38% (n=13) were younger than 30 years. Increased consumption frequency was reported for all 4 substances and most alarming with mephedrone (48%).

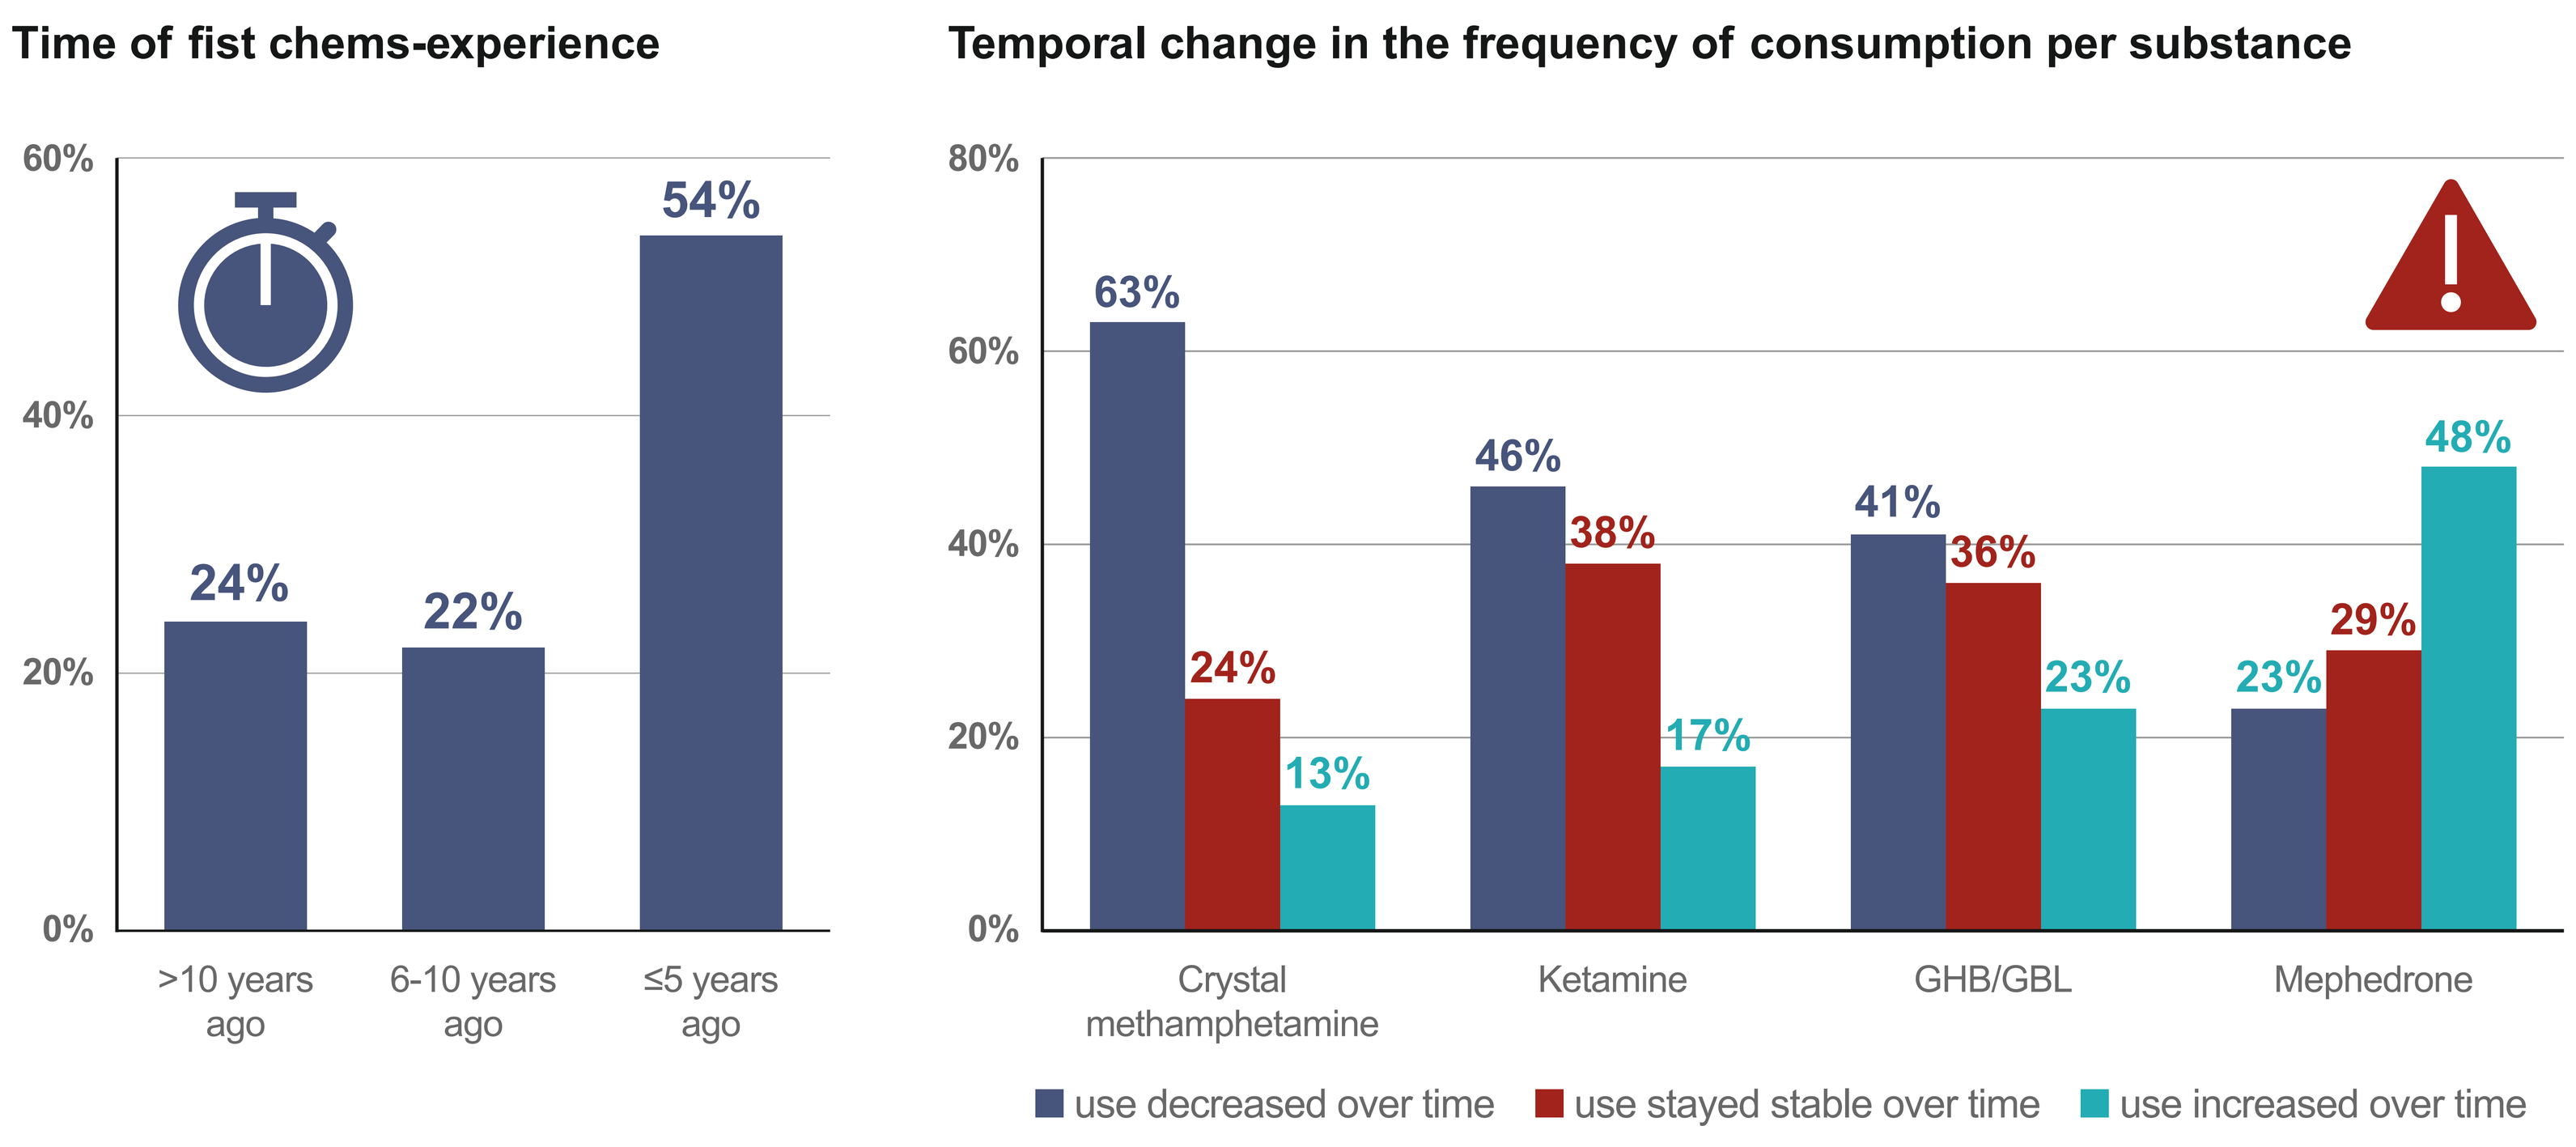
Time of fist chems-experience
60%
40%
20%
0%
24%
22%
54%
>10 years ago
6-10 years ago
≤5 years ago
Temporal change in the frequency of consumption per substance
80%
60%
40%
20%
0%
63% 24% 13%
46% 38% 17%
41% 36% 23%
23% 29% 48%
Crystal methamphetamine
Ketamine
GHB/GBL
Mephedrone
use decreased over time
use stayed stable over time
use increased over time

Figure 7. Self-assessment of chemsex burden

62% report to have their consumption behavior under control. 23% state that they sometimes to often feel loss of control regarding their chems consumption. 12% take or have already taken professional help, e.g. counselling or therapy.

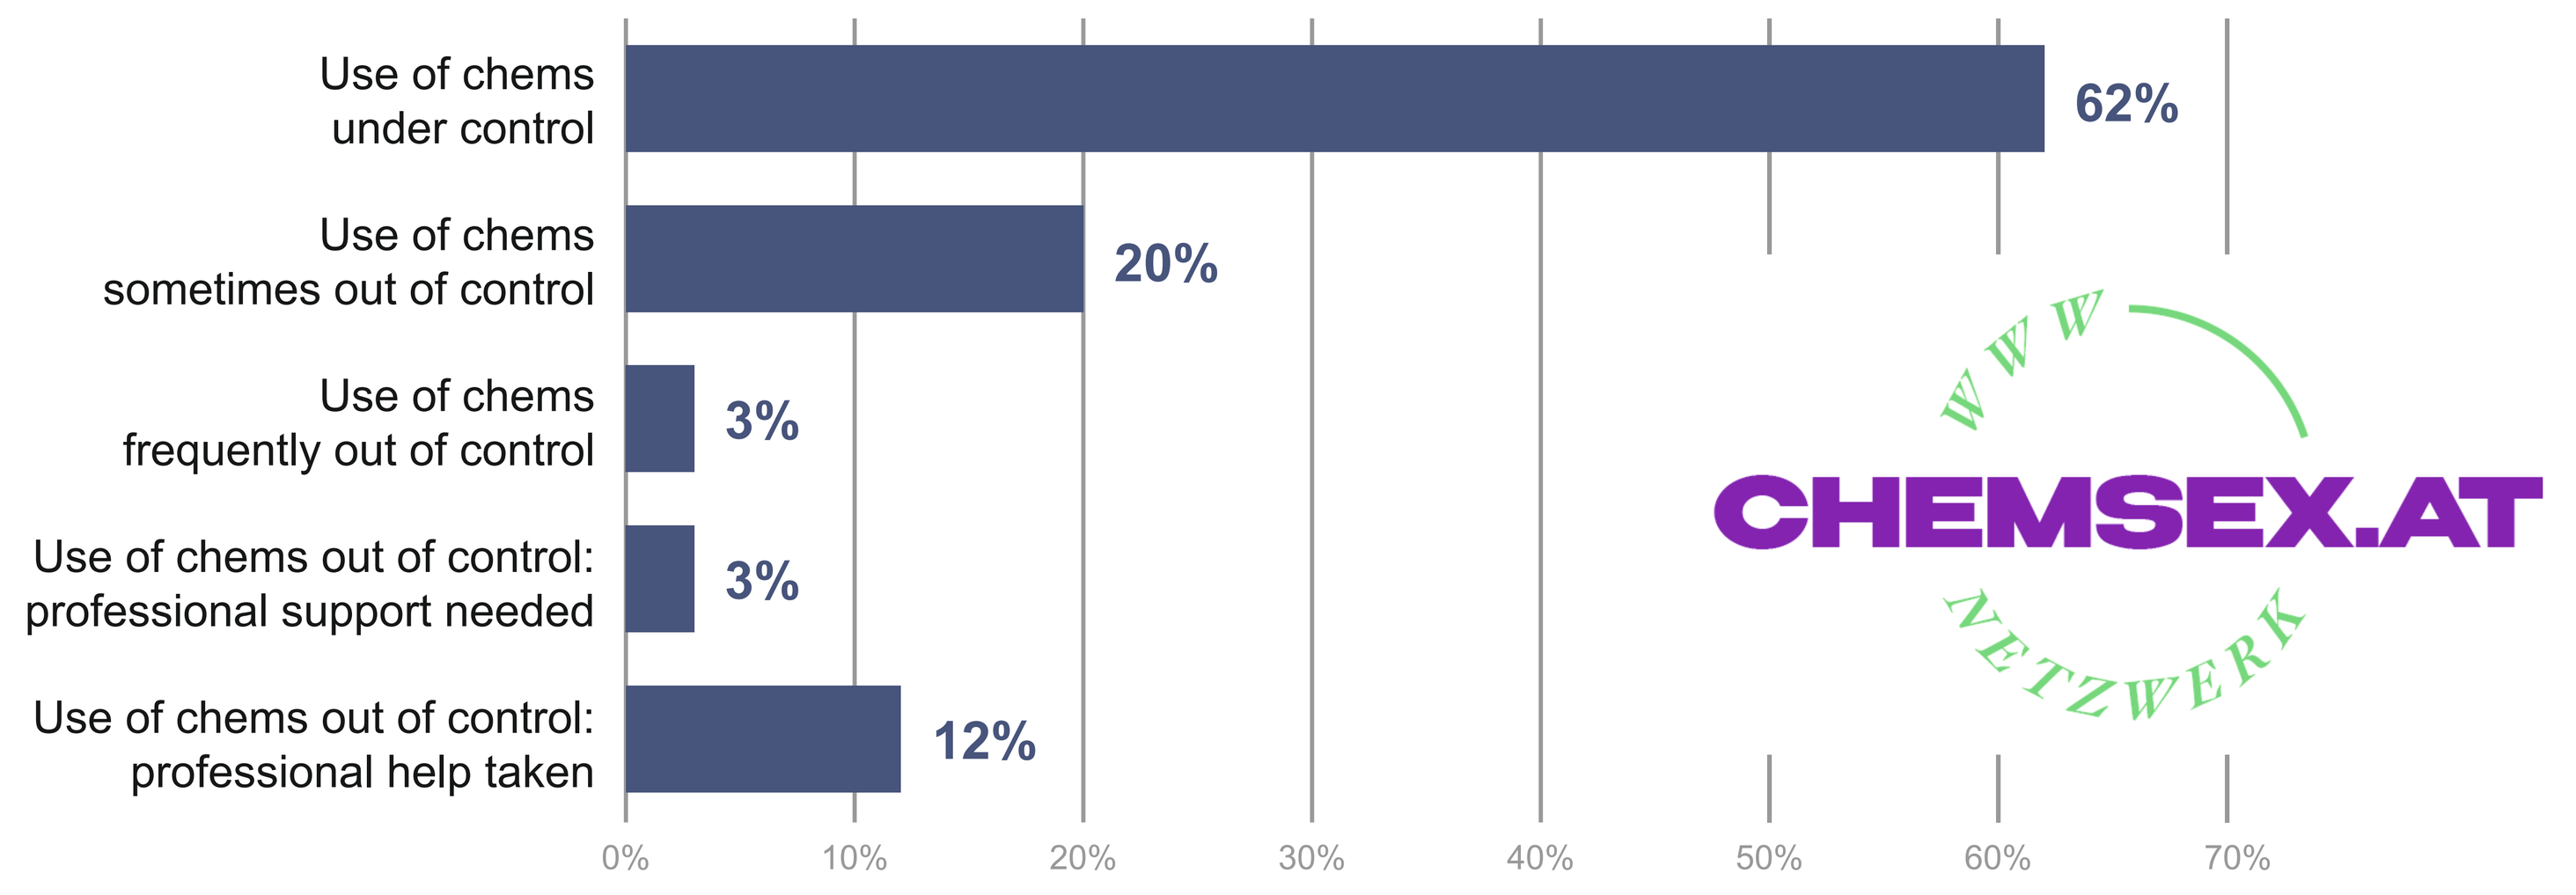
Use of chems under control 62%
Use of chems sometimes out of control 20%
Use of chems frequently out of control 3%
Use of chems out of control: professional support needed 3%
Use of chems out of control: professional help taken 12%
0% 10% 20% 30% 40% 50% 60% 70%
WWW
CHEMSEX.AT
NETZWERK

References: **1.** Stuart D. *Drugs and Alcohol Today*. 2019:19(1):3-10. **2.** Stevens O, et al. *Sex Health*. 2018:15(2):108-15. **3.** European MSM Internet Survey EMIS-2017 – Länderbericht für Österreich: https://aids.at/wp-content/uploads/2021/03/EMIS-2017_Bericht_Oesterreich-_Stand15032021_Final.pdf (last access: March 13, 2025). **4.** Austrian Chemsex Network: www.chemsex.at (last access: March 13, 2025). **5.** Baumgartner T. *Infection.* 2021:49(Suppl 1):50. **6.** Baumgartner T. *HIV Med.* 2023.24(Suppl 1):36.

Abbreviations: **GHB/GBL:** gamma-hydroxybutyric acid/gamma-butyrolactone; **MSM:** men who have sex with men

Acknowledgments:
We thank all volunteers participating in this survey initiated and conducted by members of the Austrian Chemsex Network (www.chemsex.at) and supported by Gilead Sciences GesmbH, Wagramer Straße 19, A-1220 Wien, Austria.

Correspondence:
Mag.ª Birgit Leichsenring, leichsenring@med-info.at

German-Austrian AIDS Congress (DÖAK), March 20-22 2025, Vienna, Austria